ACCG-2671 Demonstrates Promising Efficacy and Pharmacokinetics in a Proof-of-Concept Study in Obese NHPs, Supporting Further Development as Mono- and Combination Therapy

Xiankang Fang, Jinqiang Zhang, Nan Hu, Xinglong Jiang, Xichen Lin, Fang Zhang

Structure Therapeutics, South San Francisco, CA, USA

#3061-LB

Introduction

Amylin and GLP-1 receptor agonists reduce food intake, slow gastric emptying, and suppress glucagon. While GLP-1 agonists increase insulin secretion, amylin analogs also promote satiety and enhance insulin action, indicating in combination they can enhance efficacy over single agents for obesity treatment.[1]

Here we evaluated the pharmacokinetics (PK) and efficacy properties of a novel, small molecule dual amylin and calcitonin receptor agonist (DACRA), ACCG-2671 in obese non-human primates (NHPs). We further assessed whether efficacy of ACCG-2671 could be enhanced when combined with the GLP-1 receptor agonist aleniglipron.

Proposed physiological effects of amylin receptor activation[2, 3, 4]

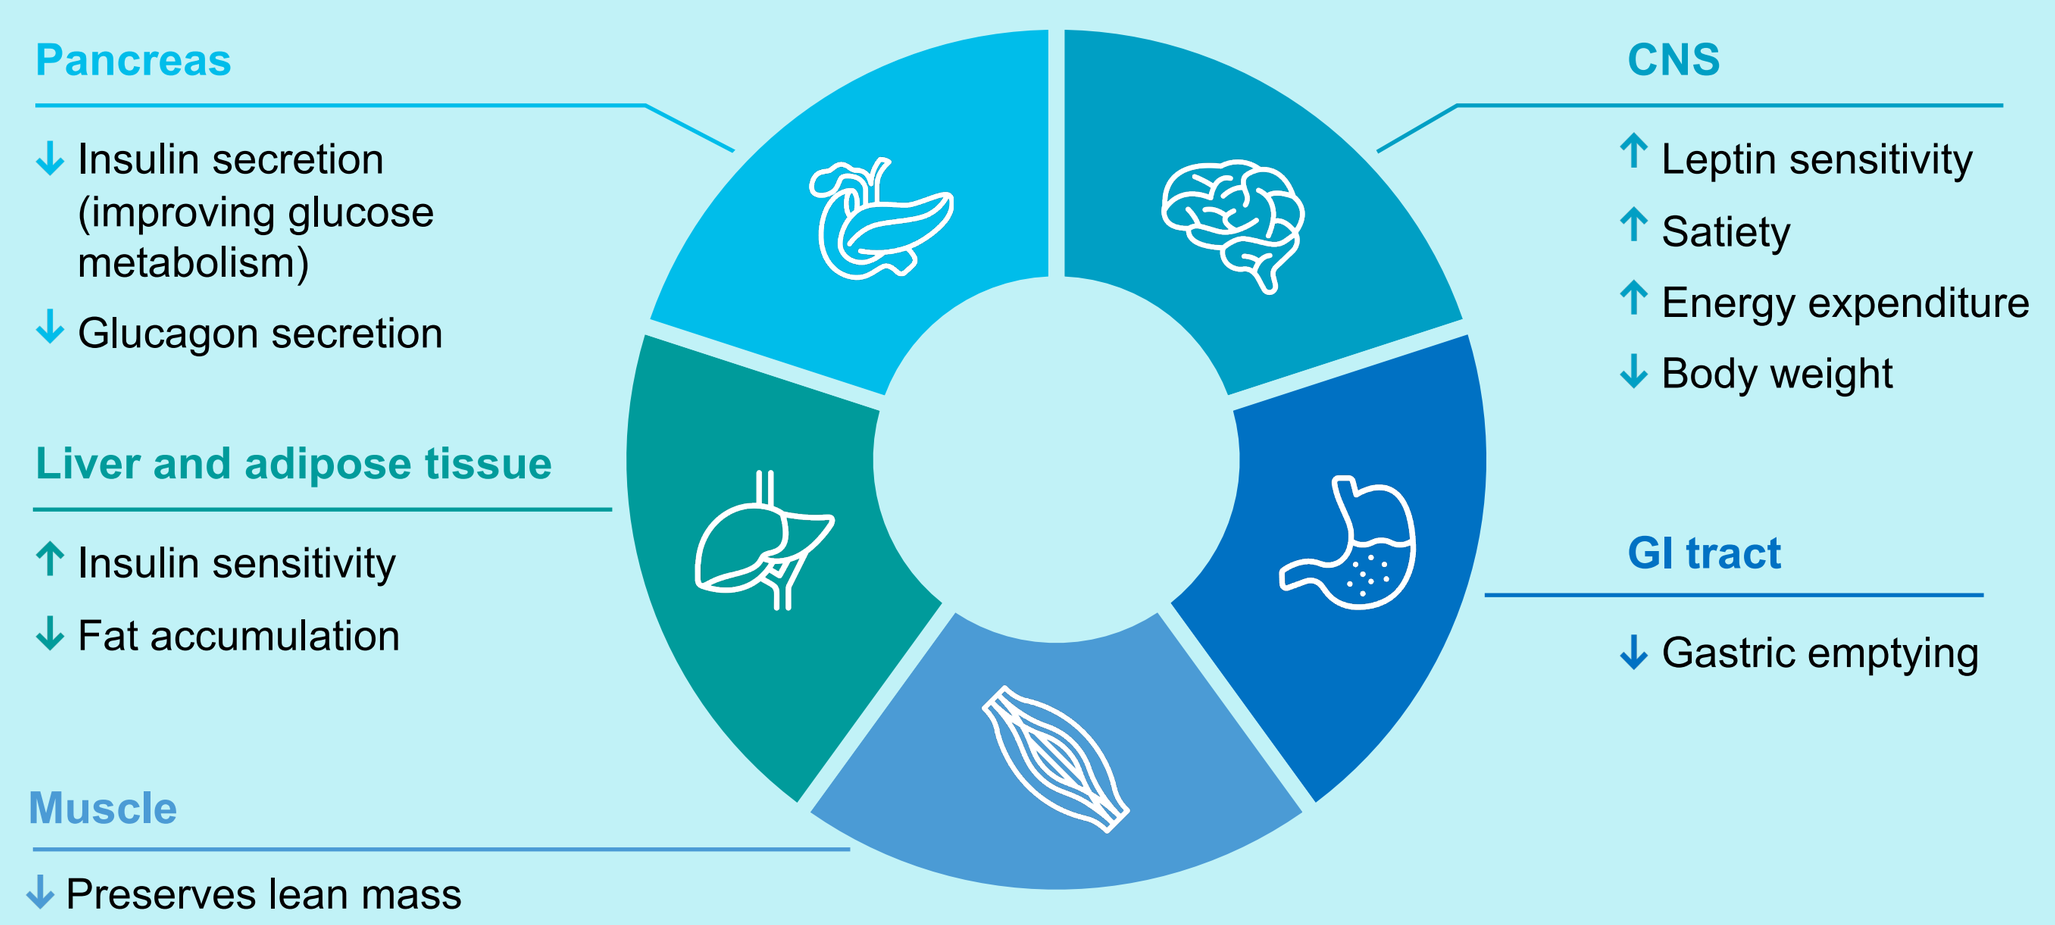

Pancreas
↓ Insulin secretion (improving glucose metabolism)
↓ Glucagon secretion
Liver and adipose tissue
↑ Insulin sensitivity
↓ Fat accumulation
Muscle
↓ Preserves lean mass
CNS
↑ Leptin sensitivity
↑ Satiety
↑ Energy expenditure
↓ Body weight
GI tract
↓ Gastric emptying

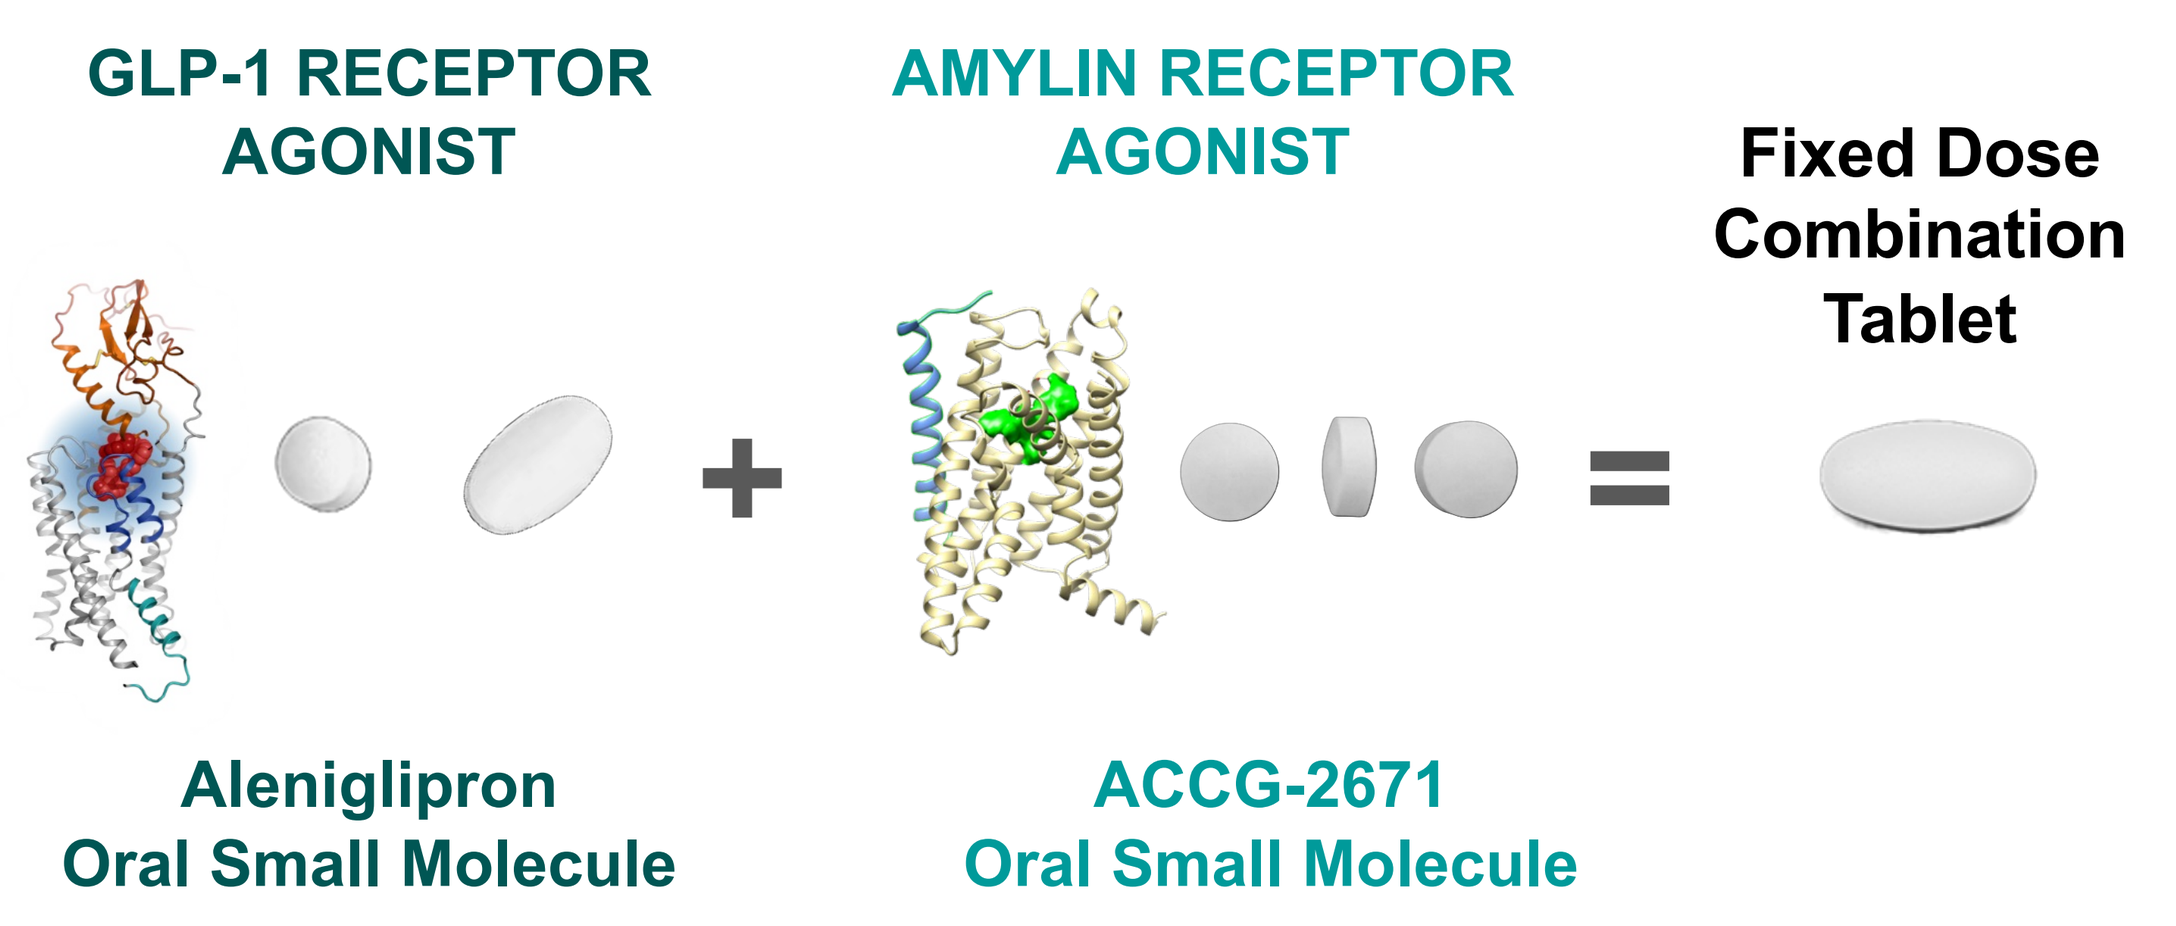

GLP-1 RECEPTOR AGONIST
AMYLIN RECEPTOR AGONIST
Fixed Dose Combination Tablet
+
=
Aleniglipron
Oral Small Molecule
ACCG-2671
Oral Small Molecule

Methods

- Obese male cynomolgus monkeys (body weight ≥8 kg, age ≥10 years, body fat ≥30%) were individually housed for the duration of the study. Animals were randomized into treatment groups (n=5 per group) based on body weight. Following a two-week dosing acclimation period, animals were treated according to the study design
 - **Monotreatment:** Obese NHPs received ACCG-2671 (0.025, 0.05, or 0.1 mg/kg, p.o., QD) to evaluate dose-dependent effects on food intake and body weight
 - **Combination treatment**: ACCG-2671 (0.05 mg/kg, p.o., QD) and aleniglipron (0.5 mg/kg, p.o., QD) were co-administered at submaximal doses, selected due to the limited body weight reduction window in obese NHPs. Effects on food intake and body weight were evaluated. Co-administration of semaglutide (1.5 nmol/kg, s.c., QD) and cagrilintide (0.5 nmol/kg, s.c., QD) was included as comparators
- Daily food intake and body weight (measured every 2 days) were recorded. Cumulative food intake and percent body weight change from baseline were analyzed
- Data are presented as mean ± SEM. Statistical significance was assessed using one-way ANOVA followed by Dunnett's multiple comparisons test

Study design: Treatment in obese NHPs

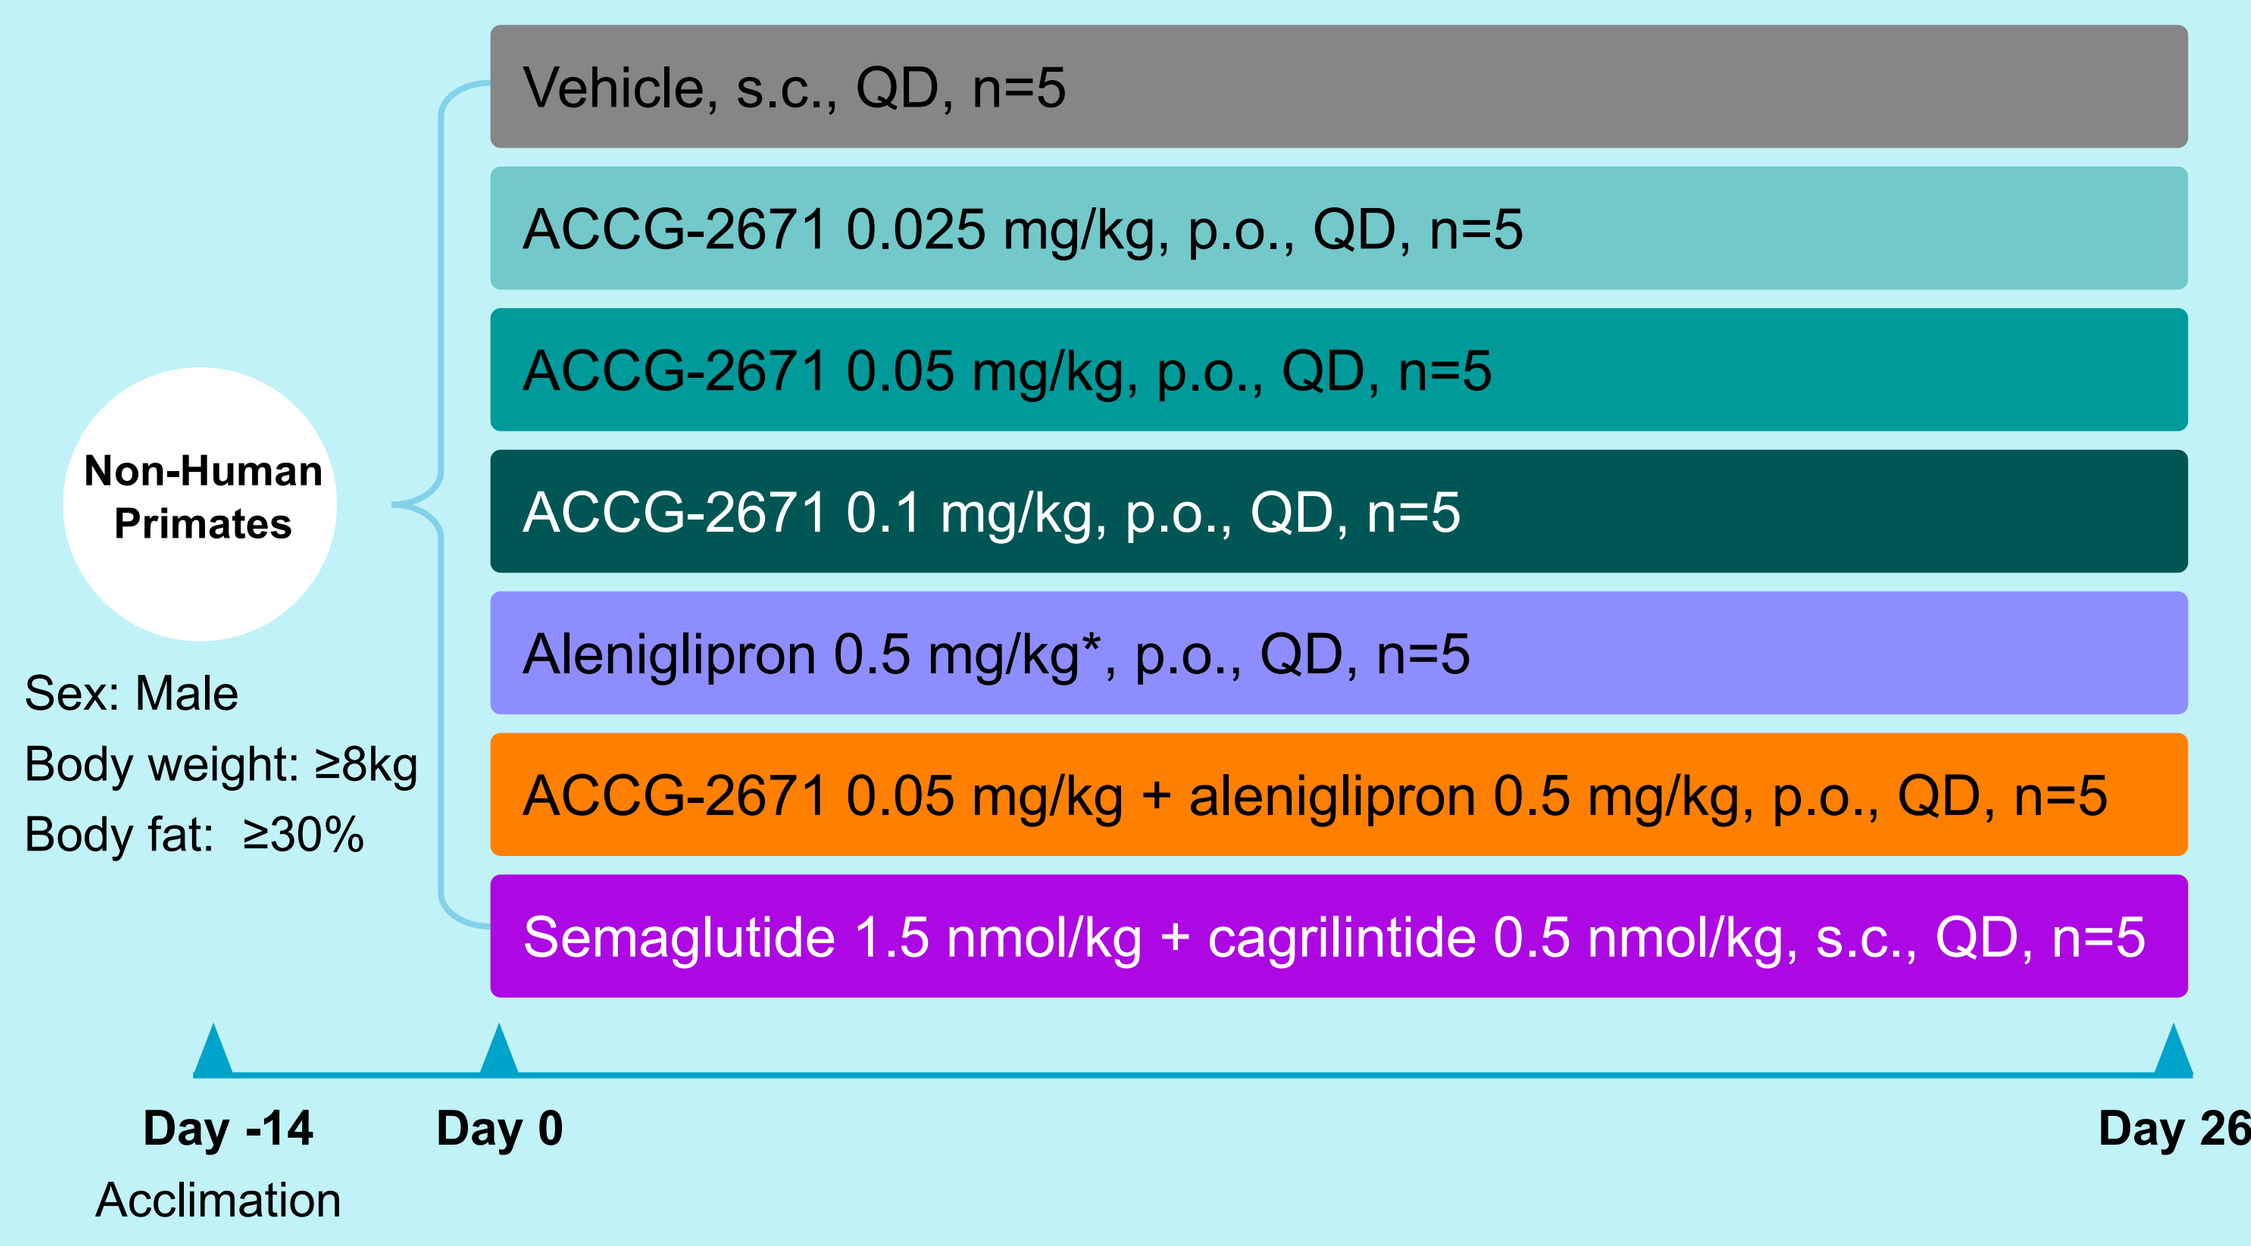

Non-Human Primates
Sex: Male
Body weight: ≥8kg
Body fat: ≥30%
Vehicle, s.c., QD, n=5
ACCG-2671 0.025 mg/kg, p.o., QD, n=5
ACCG-2671 0.05 mg/kg, p.o., QD, n=5
ACCG-2671 0.1 mg/kg, p.o., QD, n=5
Aleniglipron 0.5 mg/kg*, p.o., QD, n=5
ACCG-2671 0.05 mg/kg + aleniglipron 0.5 mg/kg, p.o., QD, n=5
Semaglutide 1.5 nmol/kg + cagrilintide 0.5 nmol/kg, s.c., QD, n=5
Day -14
Acclimation
Day 0
Day 26

*Aleniglipron was initially administered at 0.3 mg/kg for 5 days before the start of the study

Monotreatment in obese NHPs

Cumulative food intake: ACCG-2671 monotreatments

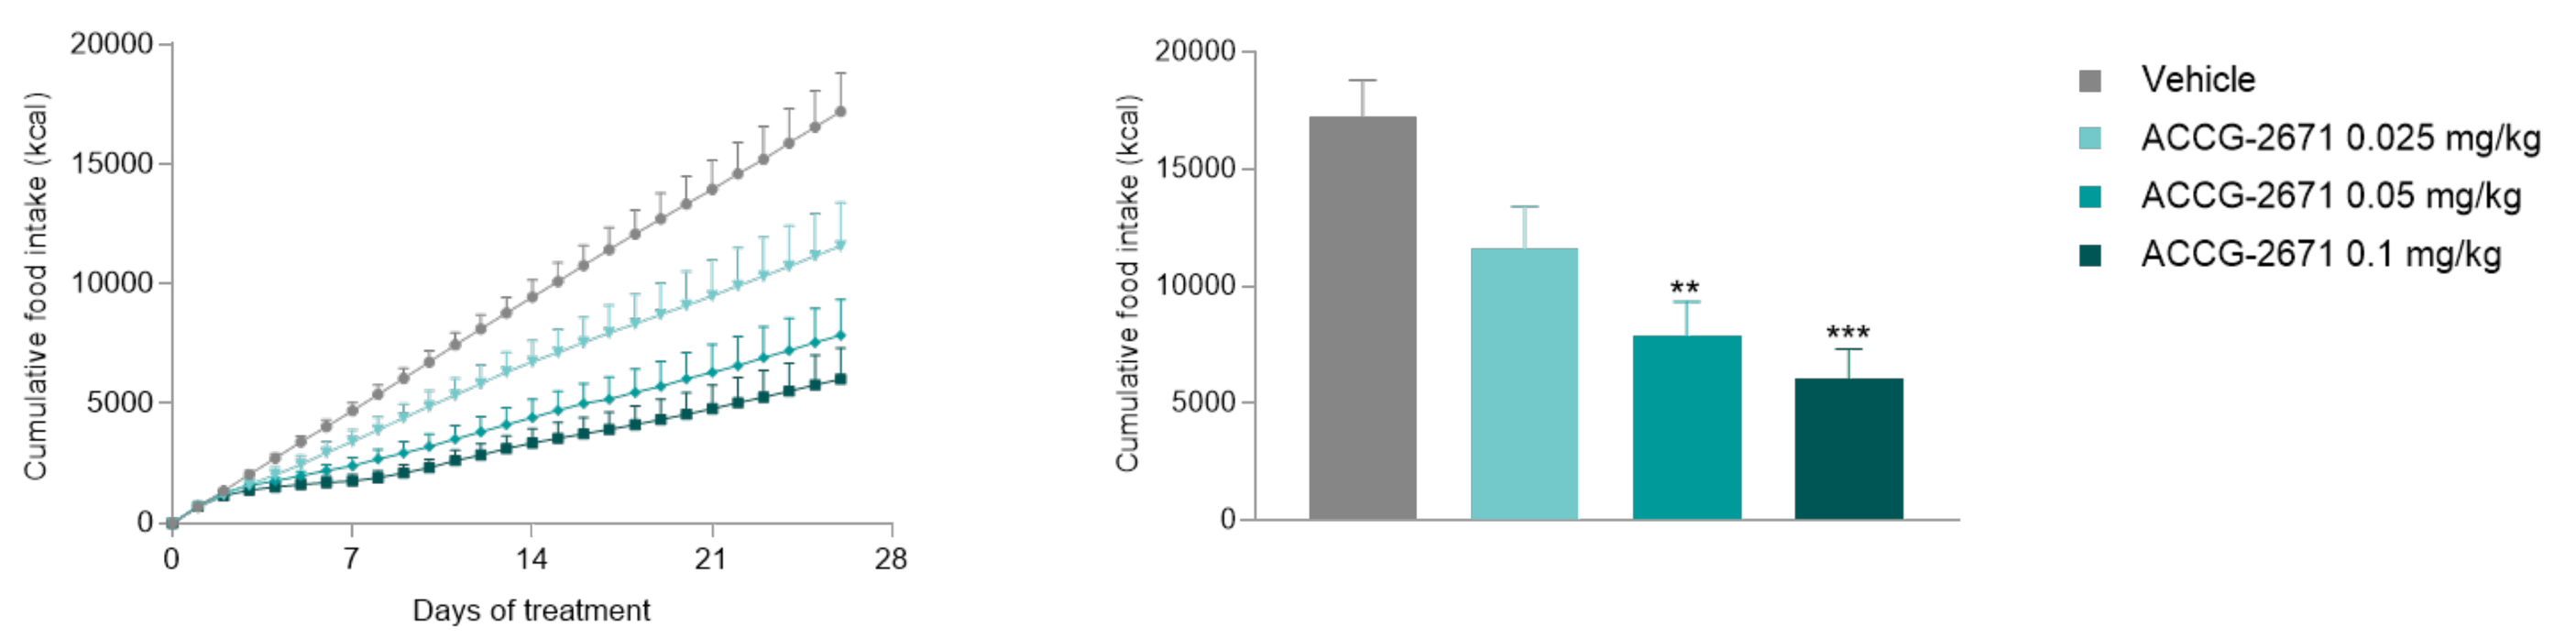

Cumulative food intake (kcal)
Days of treatment
Vehicle
ACCG-2671 0.025 mg/kg
ACCG-2671 0.05 mg/kg
ACCG-2671 0.1 mg/kg
**

p<0.01, *p<0.001, vs Vehicle group by one-way ANOVA test

ACCG-2671 oral daily dosing produced a dose-dependent reduction in cumulative food intake in obese NHPs

Chronic body weight loss: ACCG-2671 monotreatments

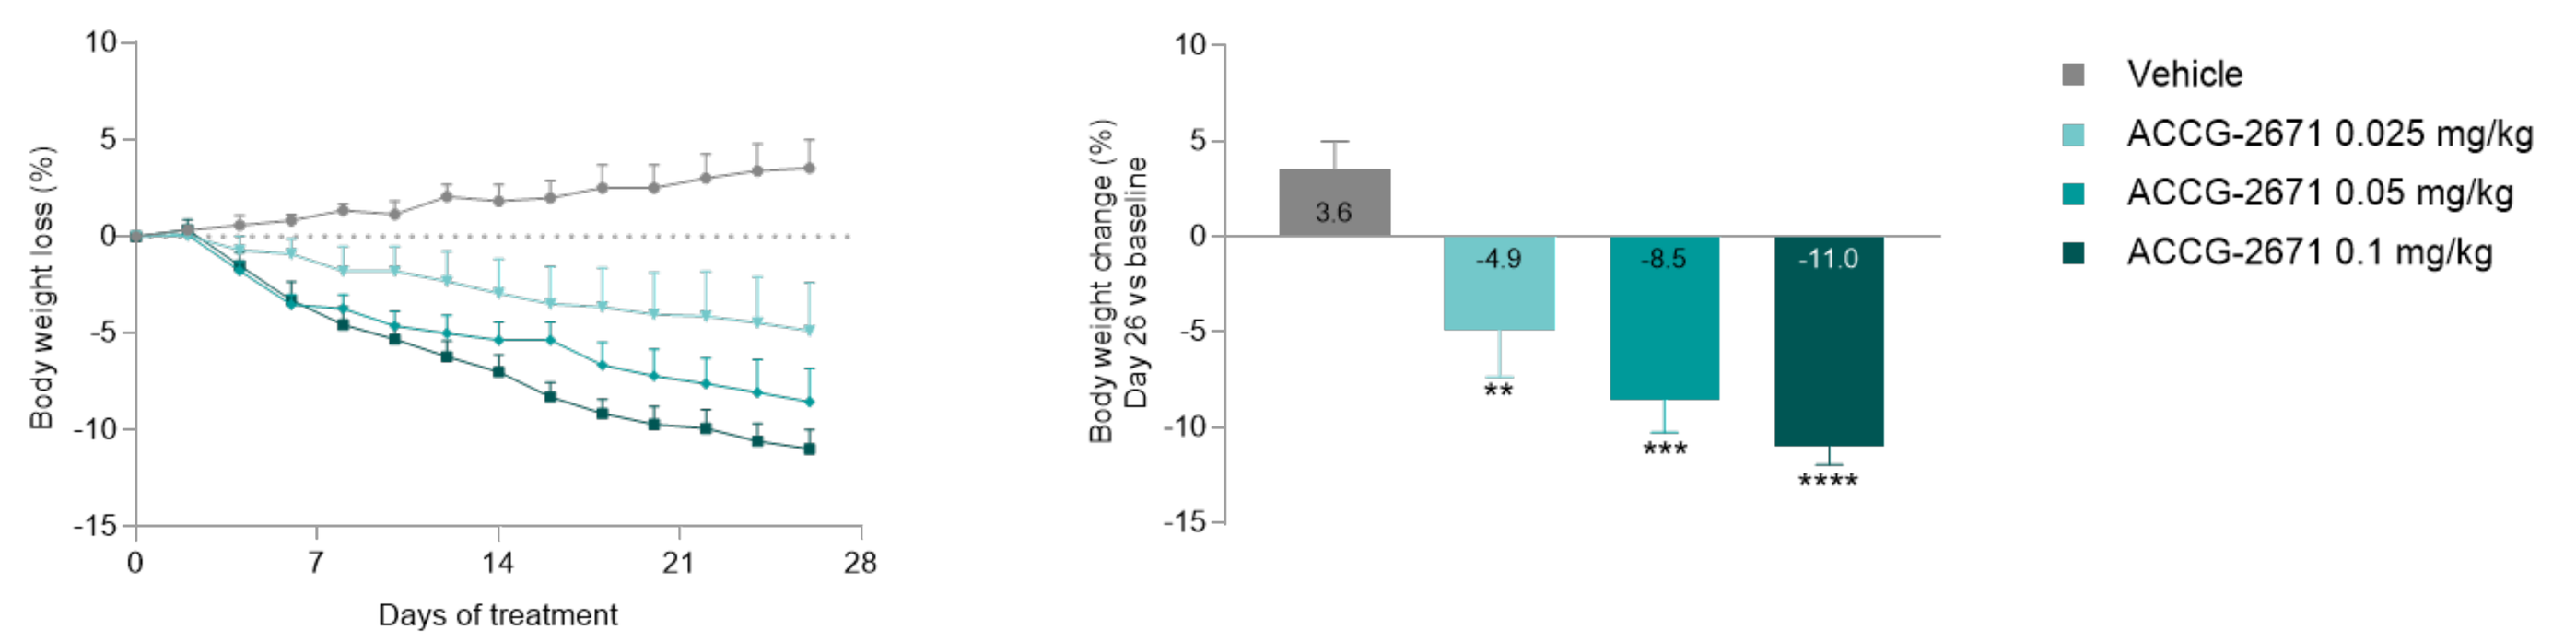

Body weight loss (%)
Days of treatment
Body weight change (%) Day 26 vs baseline
3.6
-4.9
-8.5
-11.0
**

Vehicle
ACCG-2671 0.025 mg/kg
ACCG-2671 0.05 mg/kg
ACCG-2671 0.1 mg/kg

p<0.01, *p<0.001, ****p<0.0001 vs Vehicle group by one-way ANOVA test

ACCG-2671 resulted in significant dose-dependent weight loss (-11.0% at 0.1 mg/kg and -8.5% at 0.05 mg/kg)

Monotreatment and combination treatment in obese NHPs

Cumulative food intake: ACCG-2671 + aleniglipron co-administration treatment

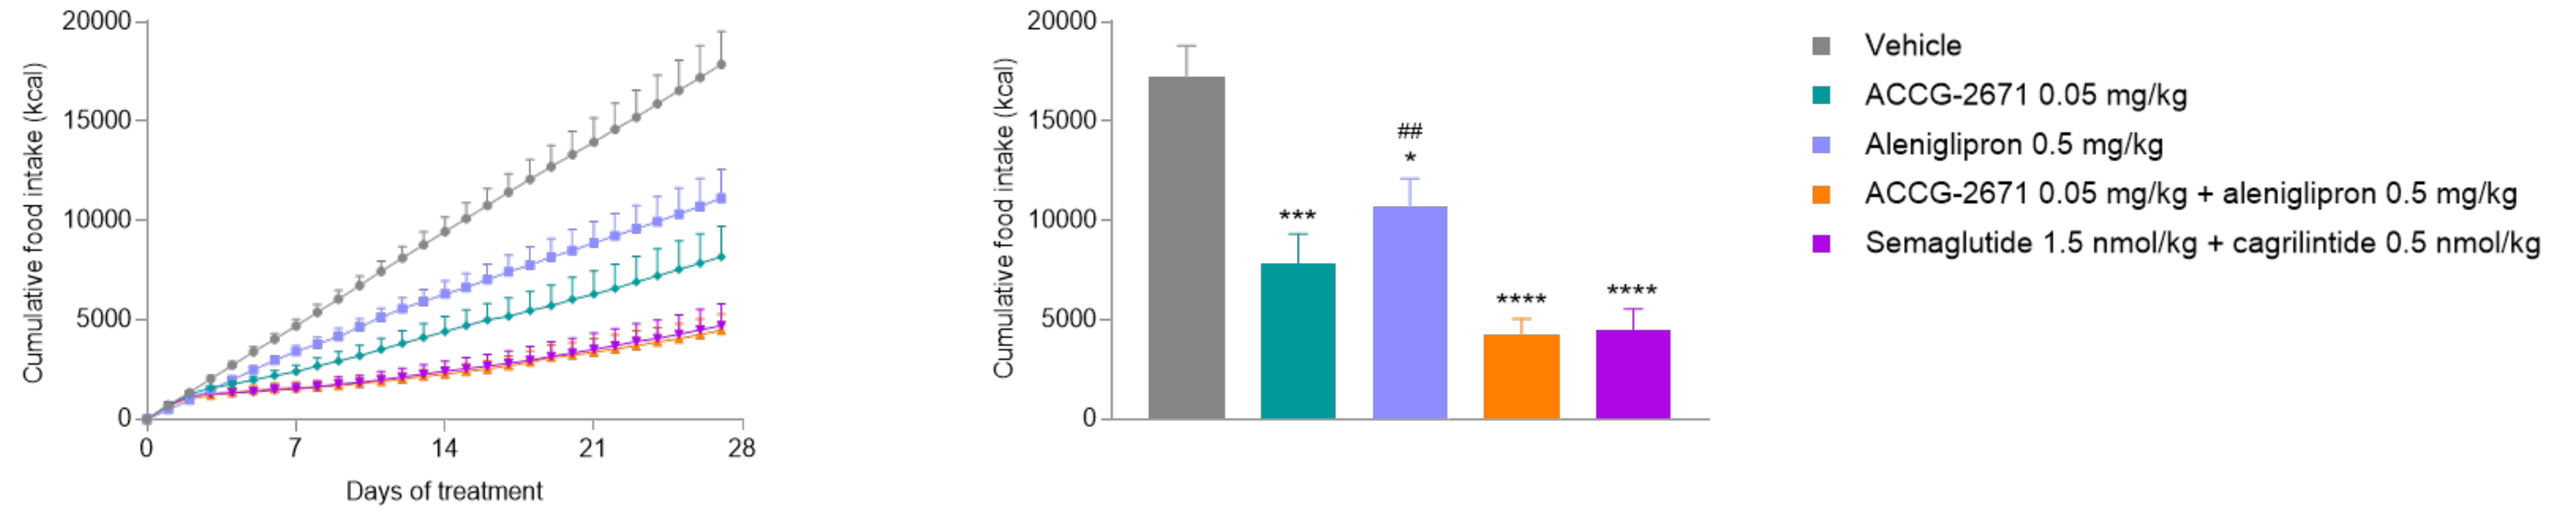

Cumulative food intake (kcal)
Days of treatment

##
*

Vehicle
ACCG-2671 0.05 mg/kg
Aleniglipron 0.5 mg/kg
ACCG-2671 0.05 mg/kg + aleniglipron 0.5 mg/kg
Semaglutide 1.5 nmol/kg + cagrilintide 0.5 nmol/kg

*p<0.05, ***p<0.001, ****p<0.0001 vs Vehicle group by one-way ANOVA test
p<0.01 vs ACCG-2671 + aleniglipron group by one-way ANOVA test

Co-administration of ACCG-2671 and aleniglipron resulted in greater reduction in food intake compared with both as monotherapies

Chronic body weight loss: ACCG-2671 + aleniglipron co-administration treatment

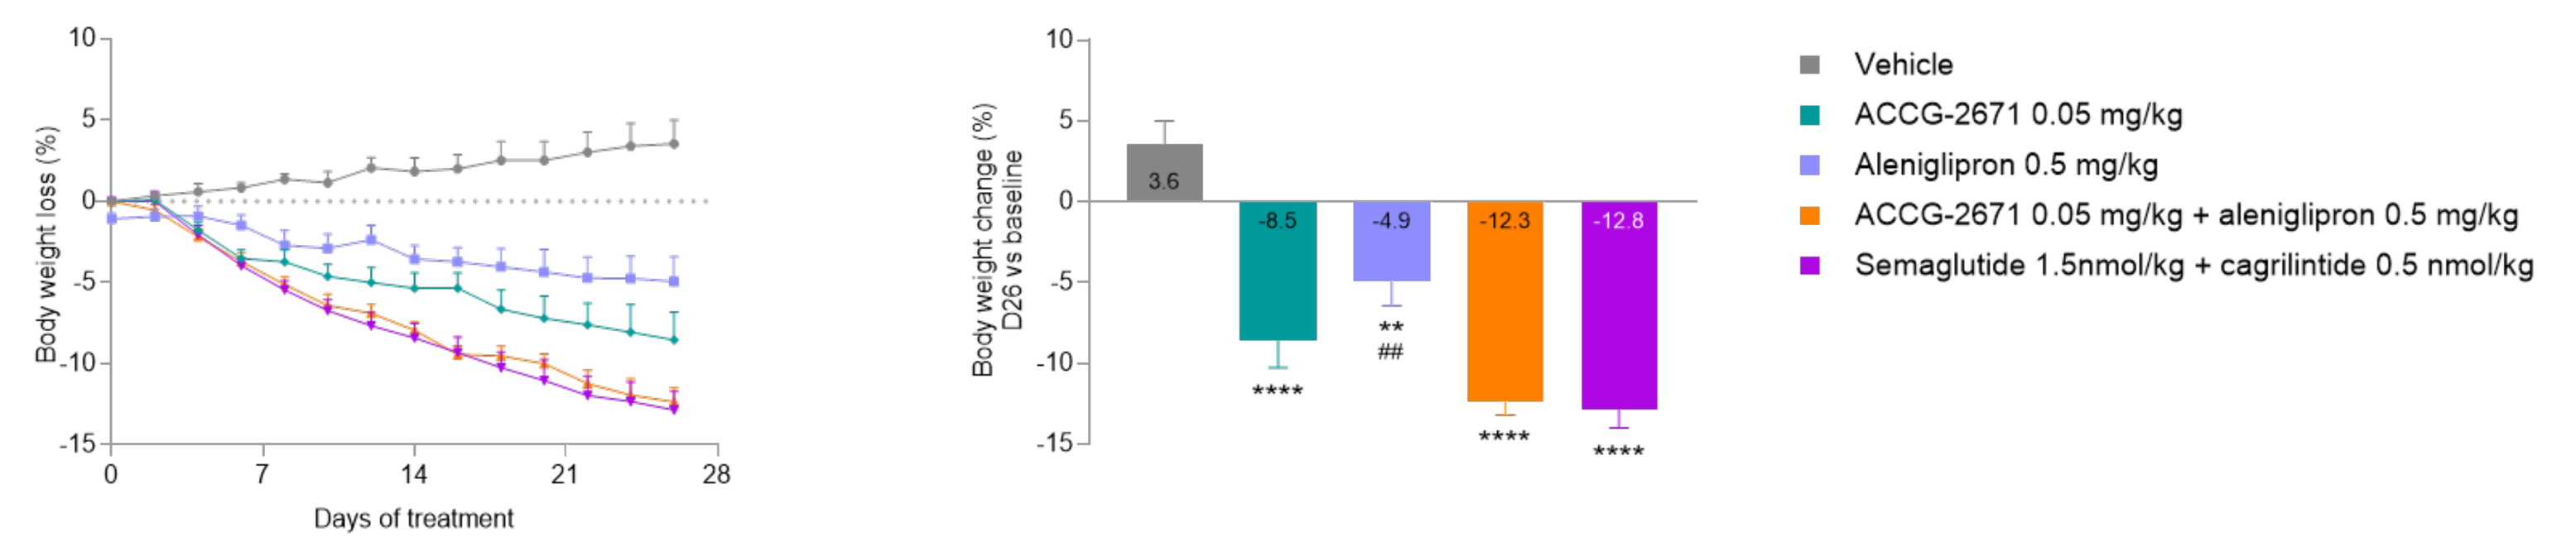

Body weight loss (%)
Days of treatment
Body weight change (%) D26 vs baseline
3.6
-8.5
-4.9
-12.3
-12.8

**
##

Vehicle
ACCG-2671 0.05 mg/kg
Aleniglipron 0.5 mg/kg
ACCG-2671 0.05 mg/kg + aleniglipron 0.5 mg/kg
Semaglutide 1.5nmol/kg + cagrilintide 0.5 nmol/kg

p<0.01, *p<0.001, ****p<0.0001 vs Vehicle group by one-way ANOVA test
p<0.01 vs ACCG-2671 + aleniglipron group by one-way ANOVA test

Co-administration of ACCG-2671 and aleniglipron resulted in −12.3% body weight reduction, greater than either monotreatment (−8.5% for ACCG-2671 0.05 mg/kg and −4.9% for aleniglipron 0.5 mg/kg)

Results

- The novel oral small molecule DACRA, ACCG-2671, demonstrated dose-dependent reductions in food intake and body weight in an obese NHP model
 - Up to 11.0% body weight loss over 26 days
- When combined with aleniglipron, ACCG-2671 resulted in greater weight loss compared to monotreatments in obese NHPs
 - Up to 12.3% body weight loss over 26 days
- ACCG-2671 yielded a long $t_{1/2}$ > 60 hr in large animals
- A Phase 1 ACCG-2671 clinical trial is currently ongoing to assess the PK profile in humans

Conclusions

- These findings support further development of ACCG-2671 as a monotherapy and potentially as an oral fixed-dose combination therapy for obesity

References

1. Volčanšek et al. *Diabetes Ther* (2025)
2. Walker et al. *Nat Rev Endocrinol* (2025)
3. Mathiesen et al. *Eur J Endocrinol* (2022)
4. Hey et al. *Pharmacol Rev* (2015)

Abbreviations:
ANOVA = analysis of variance
hr = hour
NHPs = non-human primates
PK = pharmacokinetics
QD = once daily
s.c. = subcutaneous
SEM = standard error of the mean
$t_{1/2}$ = half-life

Structure Therapeutics and the Structure Therapeutics logo are trademarks owned by the Structure Therapeutics. © 2024 Structure Therapeutics, Inc. 11/24